Utilisation of Machine Learning to Quantify Fluid Volume of Neovascular Age-Related Macular Degeneration (nARMD) Patients Based on Swept-Source Optical Coherence Tomography (SS-OCT) Imaging: The ONTARIO Study

Simrat K. Sodhi, M.Sc.[1]; Jonathan D. Oakley, Ph.D.[2]; Austin Pereira, M.D., M.Eng.[4]; Daniel B. Russakoff, Ph.D.[2] and Netan Choudhry, M.D., FRCS(C)[3,4,5]

[1] University of Cambridge, Cambridge, UK; [2] Voxeleron LLC, San Francisco, USA; [3] Vitreous Retina Macula Specialists of Toronto, Etobicoke, ON, Canada; [4] Department of Ophthalmology & Visual Sciences, University of Toronto, Toronto, ON Canada; [5] Cleveland Clinic Canada, Toronto, ON, Canada
Financial Disclosure – Netan Choudhry: Bayer (S,C,R); Novartis (S,C); Allergan (S,C); Optos (S,C,R); Carl Zeiss Meditec (C); Jonathan Oakley and Daniel Russakoff: Voxeleron (E); Simrat Sodhi and Austin Pereira (N)

PURPOSE

Evaluate the predictive ability of a deep learning-based, automated, macular fluid segmentation algorithm to determine long-term visual acuity (VA) outcomes in neovascular age-related macular degeneration (nARMD) patients using baseline swept-source optical coherence tomography (SS-OCT) and OCT-angiography (OCT-A) data.

METHODS

Twenty-two SS-OCT volumes of the macula, comprising 5,632 images from 22 nARMD subjects were used to assess retinal layer thicknesses, quantify intraretinal fluid (IRF), subretinal fluid (SRF) and fluid in serous pigment epithelium detachments (PED). Layer thicknesses were manually corrected and fluid segmentation was performed using a novel, deep learning algorithm with results validated relative to two expert graders (**Fig 1**). Seventeen treatment-naive subject eyes, from the previously reported CANADA study, were enrolled in this study. OCT-A data was used to manually define the extent of the choroidal neovascularization (CNV) in each scan (**Fig 2**). Baseline OCT morphological features and measurements were correlated using the Pearson correlation coefficient (PCC) to changes in VA to determine which features impacted the long-term visual outcomes.

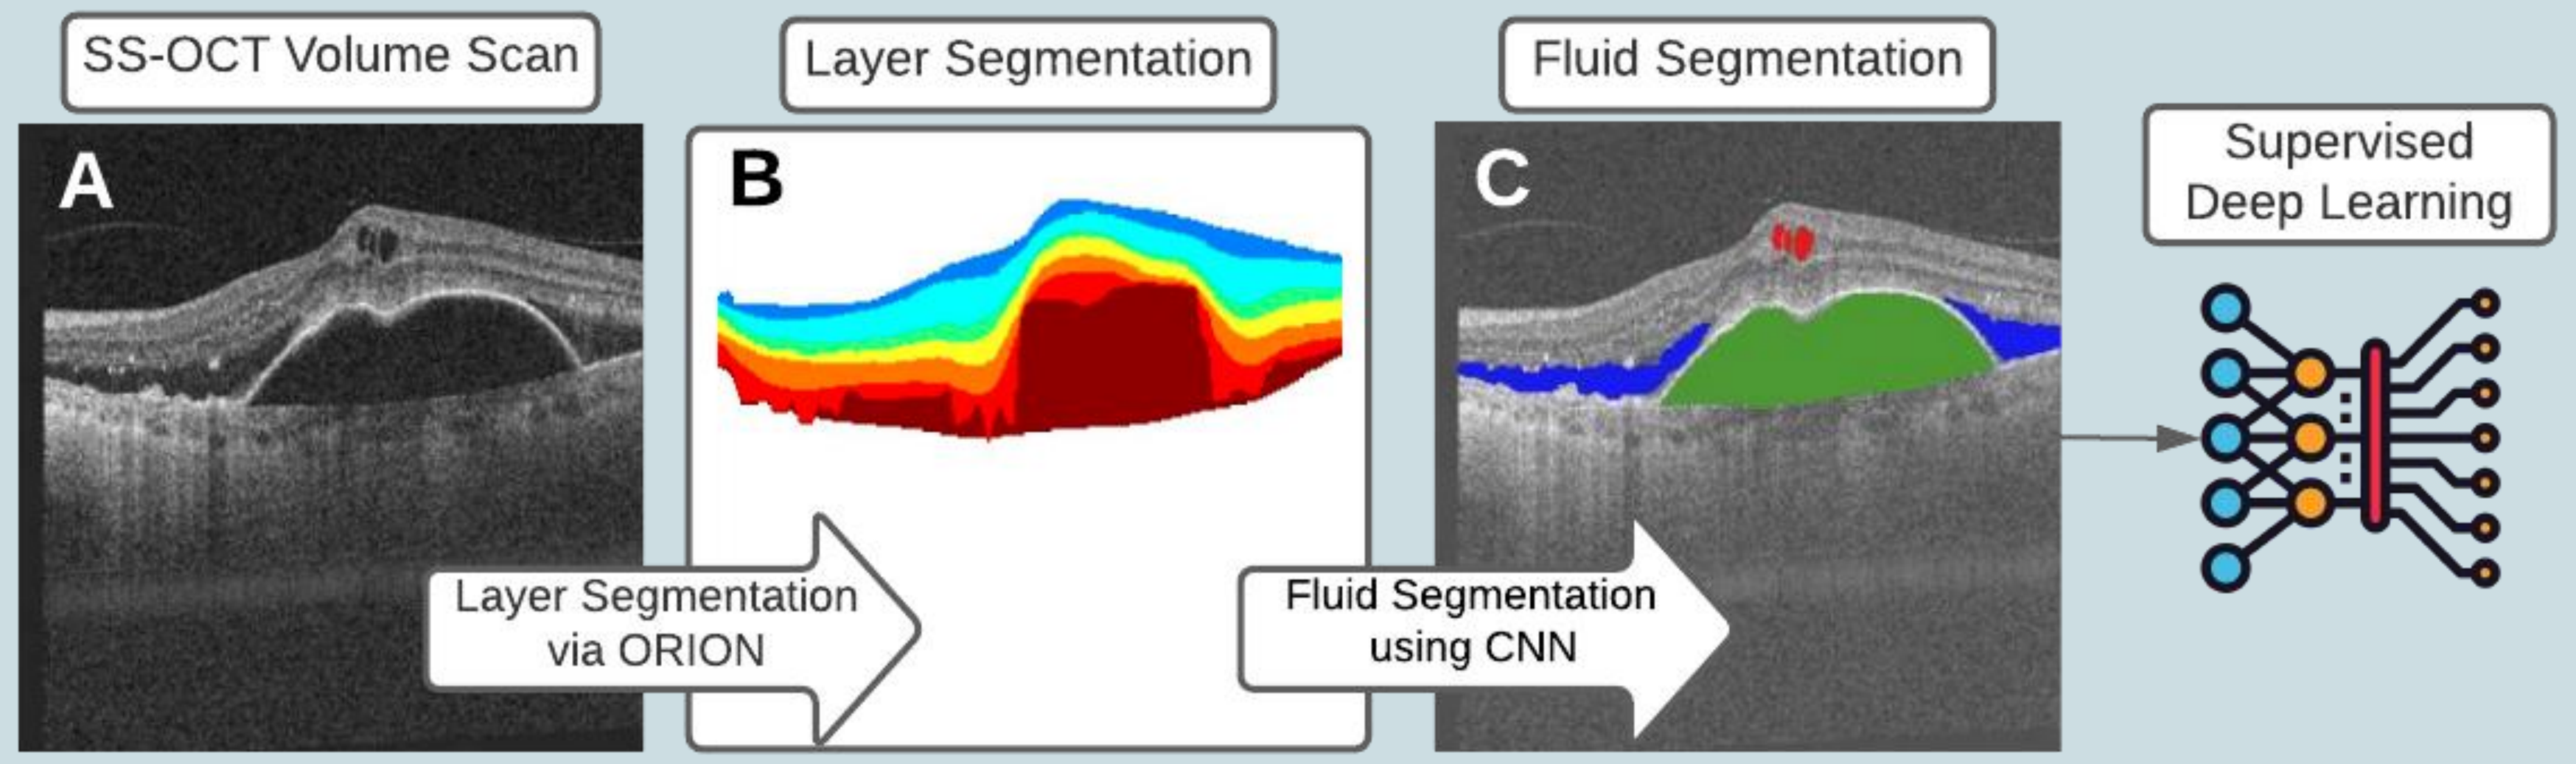
SS-OCT Volume Scan
Layer Segmentation
Fluid Segmentation
Supervised Deep Learning
A
B
C
Layer Segmentation via ORION
Fluid Segmentation using CNN

Figure 1: Each **(A)** SS-OCT volume scan is first automatically segmented using Orion into 8 retinal interfaces. This results in **(B)** 7 layers that can be encoded into image form and used as an additional channel in model creation, thus encoding spatial information regarding the location of **(C)** fluid within the retina.

RESULTS

Total retinal fluid (IRF, SRF and PED) volume at baseline had the closest correlation to improvement in BCVA at month 12 (PCC=0.652, p=0.005) (**Fig 3**). Fluid was subsequently sub-categorized into IRF, SRF and PED, with PED volume having the next highest correlation (PCC=0.648, p=0.005) to BCVA improvement. Average total retinal thickness in isolation demonstrated poor correlation (PCC=0.334, p=0.189), and mean CNVM size from 3 mm OCT-A scans showed even lower correlation to BCVA change (PCC=0.072, p=0.784). When two features were combined and correlated with visual outcomes, the highest correlation increased to PCC=0.695 (p=0.002) using mean CNVM size and total fluid volume in synchrony.

RESULTS

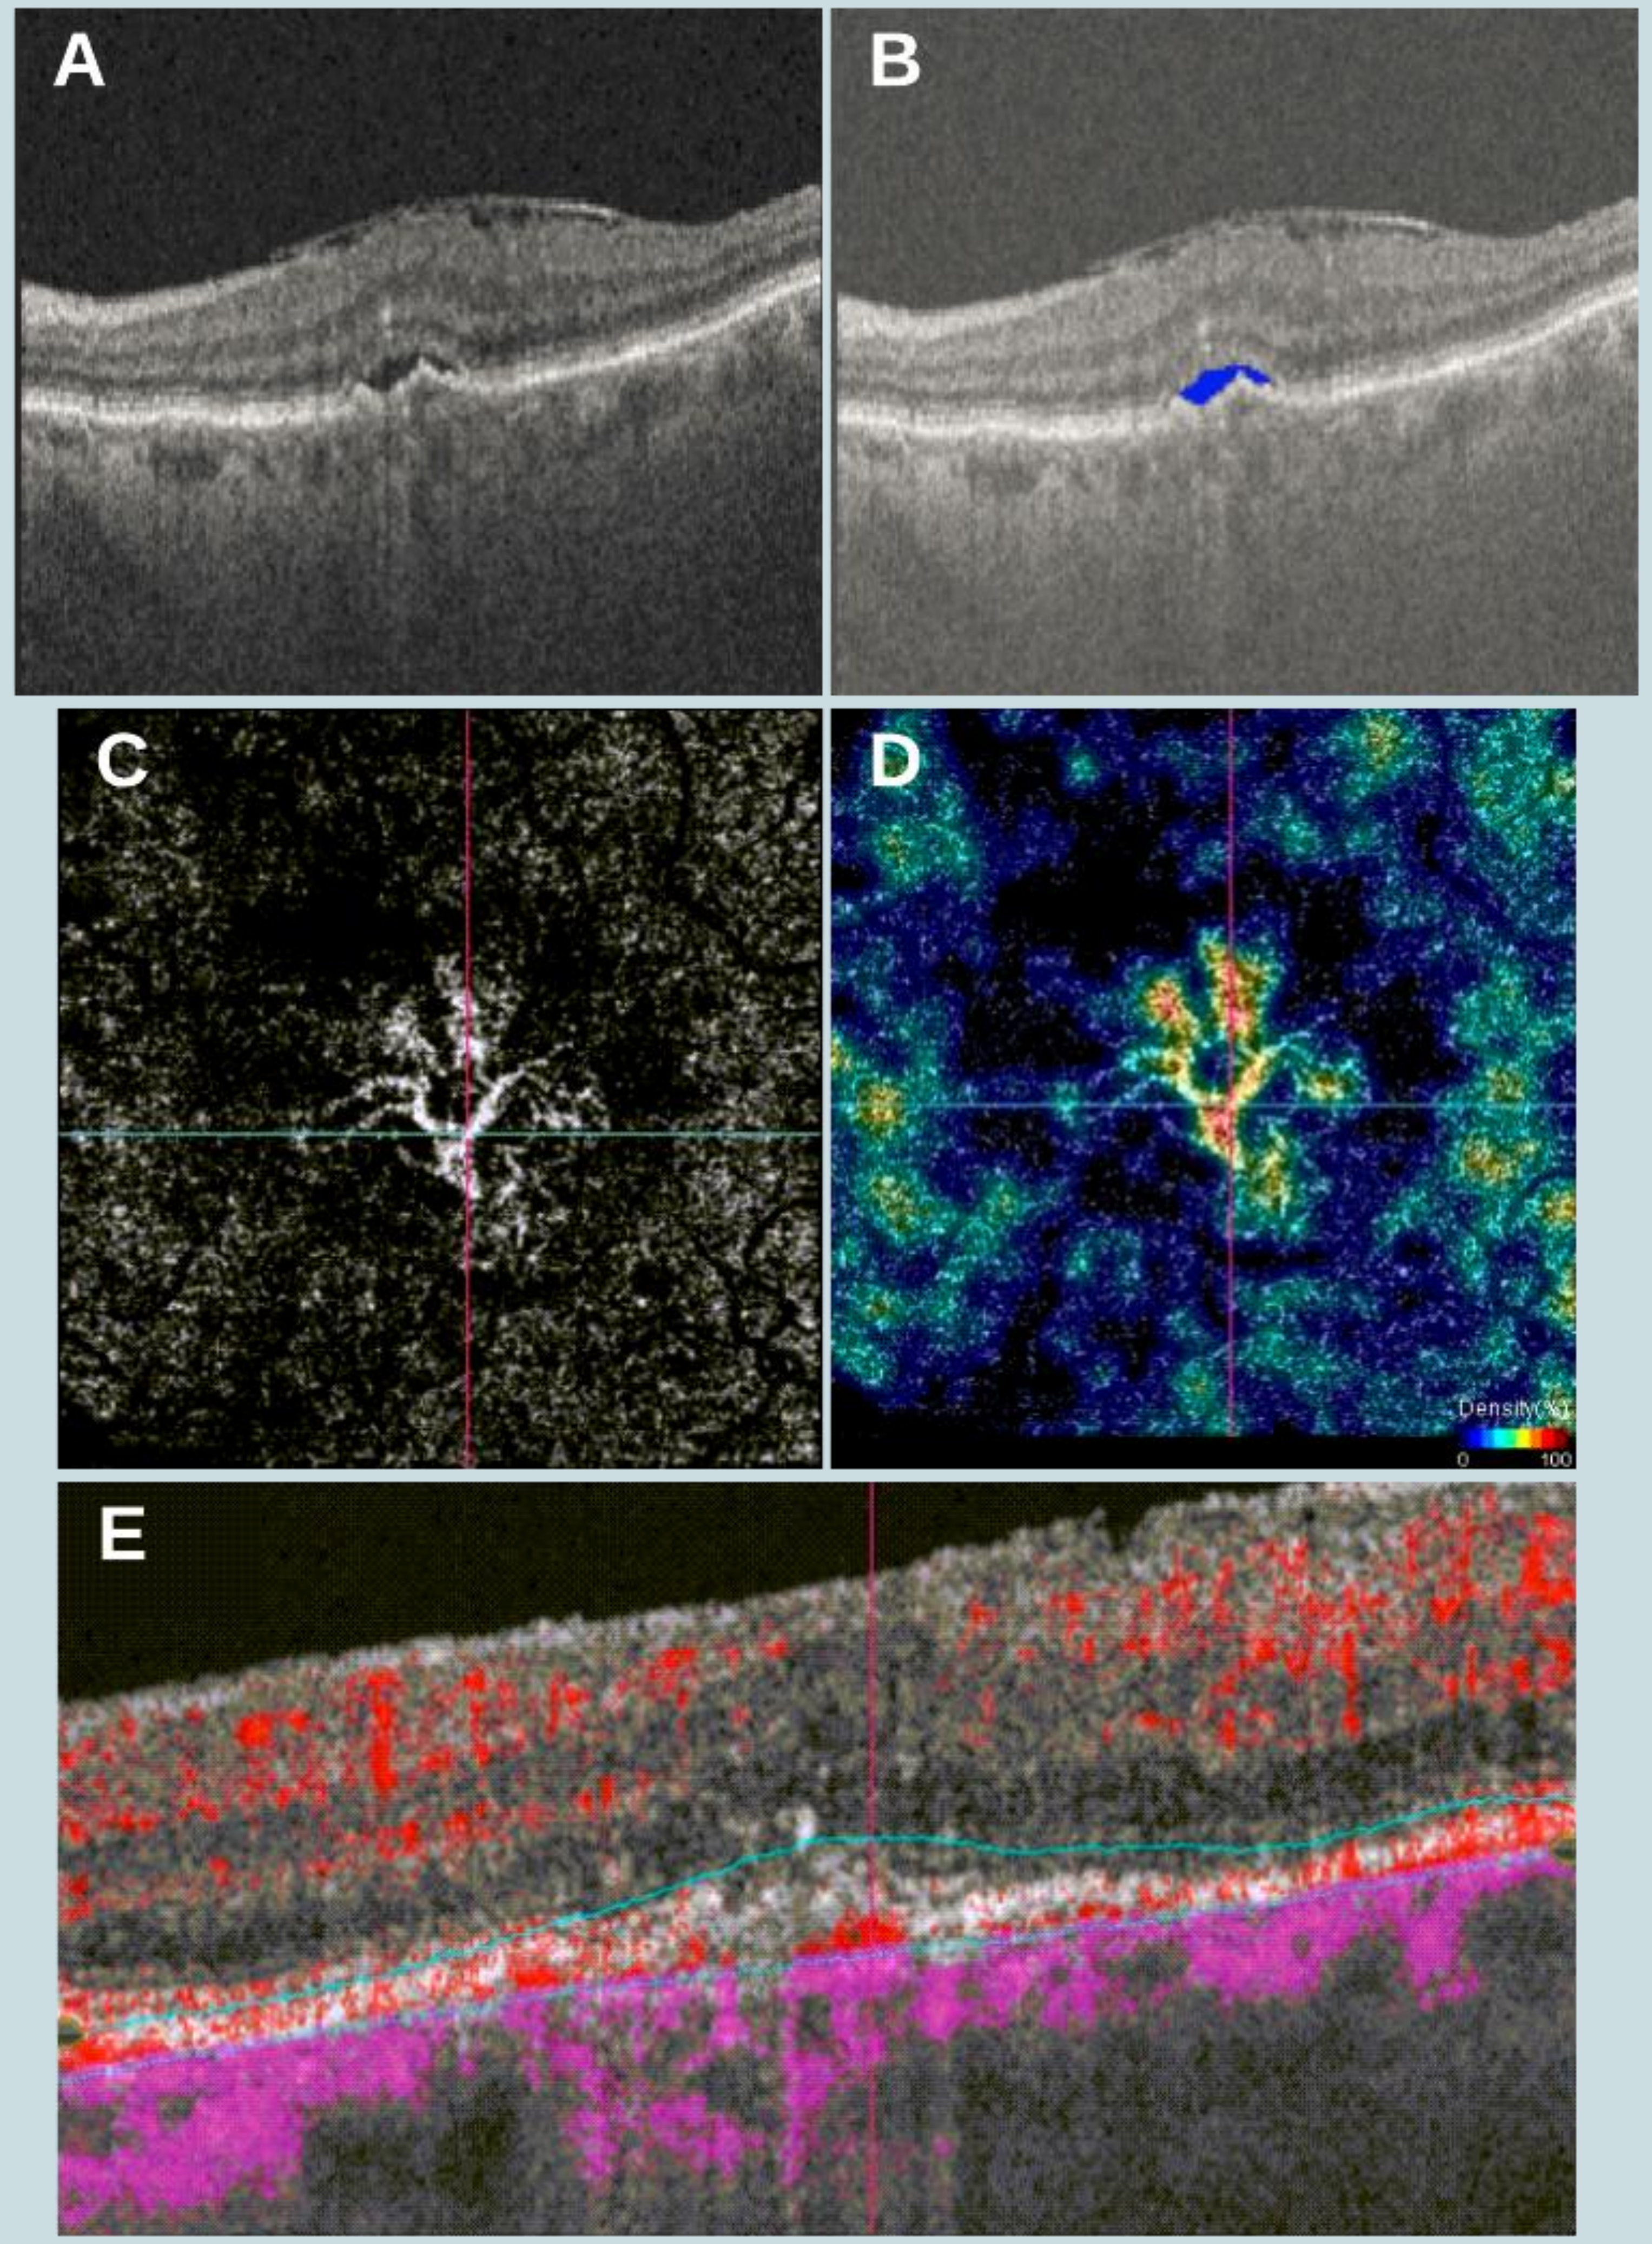
A
B
C
D
E
Density(%)
0
100

Figure 2: (A) SS-OCT volume scan of 87 year old female patient with type I CNV, SRF and epiretinal membrane (ERM); **(B)** fluid segmentation using a convolutional neural network (CNN) highlights SRF in blue; **(C)** corresponding OCT-A scan depicting segmented CNVM from outer retina (OR) slab; **(D)** density flow highlighting areas of increased flow; **(E)** corresponding flow B-scan from single horizontal B-scan through center of CNVM.

A

Feature 1	PCC	p-value
Total Fluid	0.6521	0.0046
PED	0.6481	0.0049
SRF	0.4824	0.0499
6 mm average CST	0.3344	0.1895
Density map – central – % 6 mm	0.3241	0.2045
6 mm inferior CST	0.3174	0.2145
6 mm nasal	0.3122	0.2225
6 mm temporal	0.2820	0.2728
Density map – superior – % 6 mm	0.2522	0.3288
6 mm superior	0.2108	0.4168

B

Feature 1	Feature 2	PCC	p-value
CNVM Mean size (μm²) – 3 mm OCTA	Total Fluid	0.6951	0.0099
PED	IRF	0.6752	0.0141
CNVM Mean size (μm²) – 6 mm OCTA	Total Fluid	0.6751	0.0141
CNVM Mean size (μm²) – 3 mm OCTA	PED	0.6721	0.0149
Total Fluid	SRF	0.6690	0.0157
Density map – inferior – % 6 mm	PED	0.6669	0.0163
Density map – superior – % 6 mm	Total Fluid	0.6659	0.0165
Total Fluid	IRF	0.6634	0.0172
Density map – inferior – % 6 mm	Total Fluid	0.6634	0.0172
Density map – central – % 6 mm	PED	0.6606	0.0181

Figure 3: (A) The 10 best correlating features to logMar change **(B)** the 10 best pairwise correlating features to logMar change.

CONCLUSION

In isolation, total fluid volume best correlates with change in BCVA values between baseline and week 52. In combination with complimentary information from OCT-A, an improvement in the linear correlation score was observed. Average total retinal thickness provided a lower correlation, and thus provides a lower predictive outcome than alternative metrics assessed. In this pilot study, SS-OCT and OCT-A data together correlated better to visual acuity outcomes then any one metric in isolation. Clinically, a machine-learning approach to analyzing fluid metrics in combination with lesion size may provide an advantage in personalizing therapy and predicting BCVA outcomes.

ACKNOWLEDGMENTS

The authors would like to thank John Golding, BA and Regine Maranion, RN for their assistance. This study was supported by funding from Bayer Inc.

For any questions or comments, contact simrat.k.sodhi@gmail.com